The authors report no financial conflicts of interest.

Who Needs Residential Treatment? *The Adolescent Population Admitted to a Residential Treatment Facility in New York*

Celso Gomes, PhD
cgomes@aichhorn.org

Dariany Garcia, BS
dgarcia@aichhorn.org

Michael Pawel, MD
mpawel@aichhorn.org

Piper Smith, LCSW
psmith@aichhorn.org

BACKGROUND: Residential treatment is expensive, restrictive and outcomes are poorly understood so contemporary focus is on treatment in the community. But a subset of children continues to enter residential placements. What characterizes these children? Why do some enter a Residential Treatment Facility (RTF), the most restrictive type of placement, and is this appropriate?

METHODS

1. Conducted records review of population admitted to the August Aichhorn Center RTF in New York (RTF-M).
2. Compared population characteristics with those of Residential Treatment Centers (RTC) in New York State, which are the most restrictive residential placements in the child welfare system. Assessed significance of differences using Chi Square tests.

RESULTS

1. The most common referral source for RTF-M was psychiatric hospitalization vs. home of a parent or relative for RTC admits.
2. 75% of the RTF-M population was admitted from psychiatric hospitalization or the juvenile justice system vs. 33% for the RTCs.
3. Psychotic symptoms, assaultiveness and suicidal behavior are pervasive throughout the RTF-M population.
4. The RTF-M population, which was highly screened by a State-operated office separate from the RTF-M, had markedly more adverse histories and symptomatology prior to admission than the New York State RTC population.

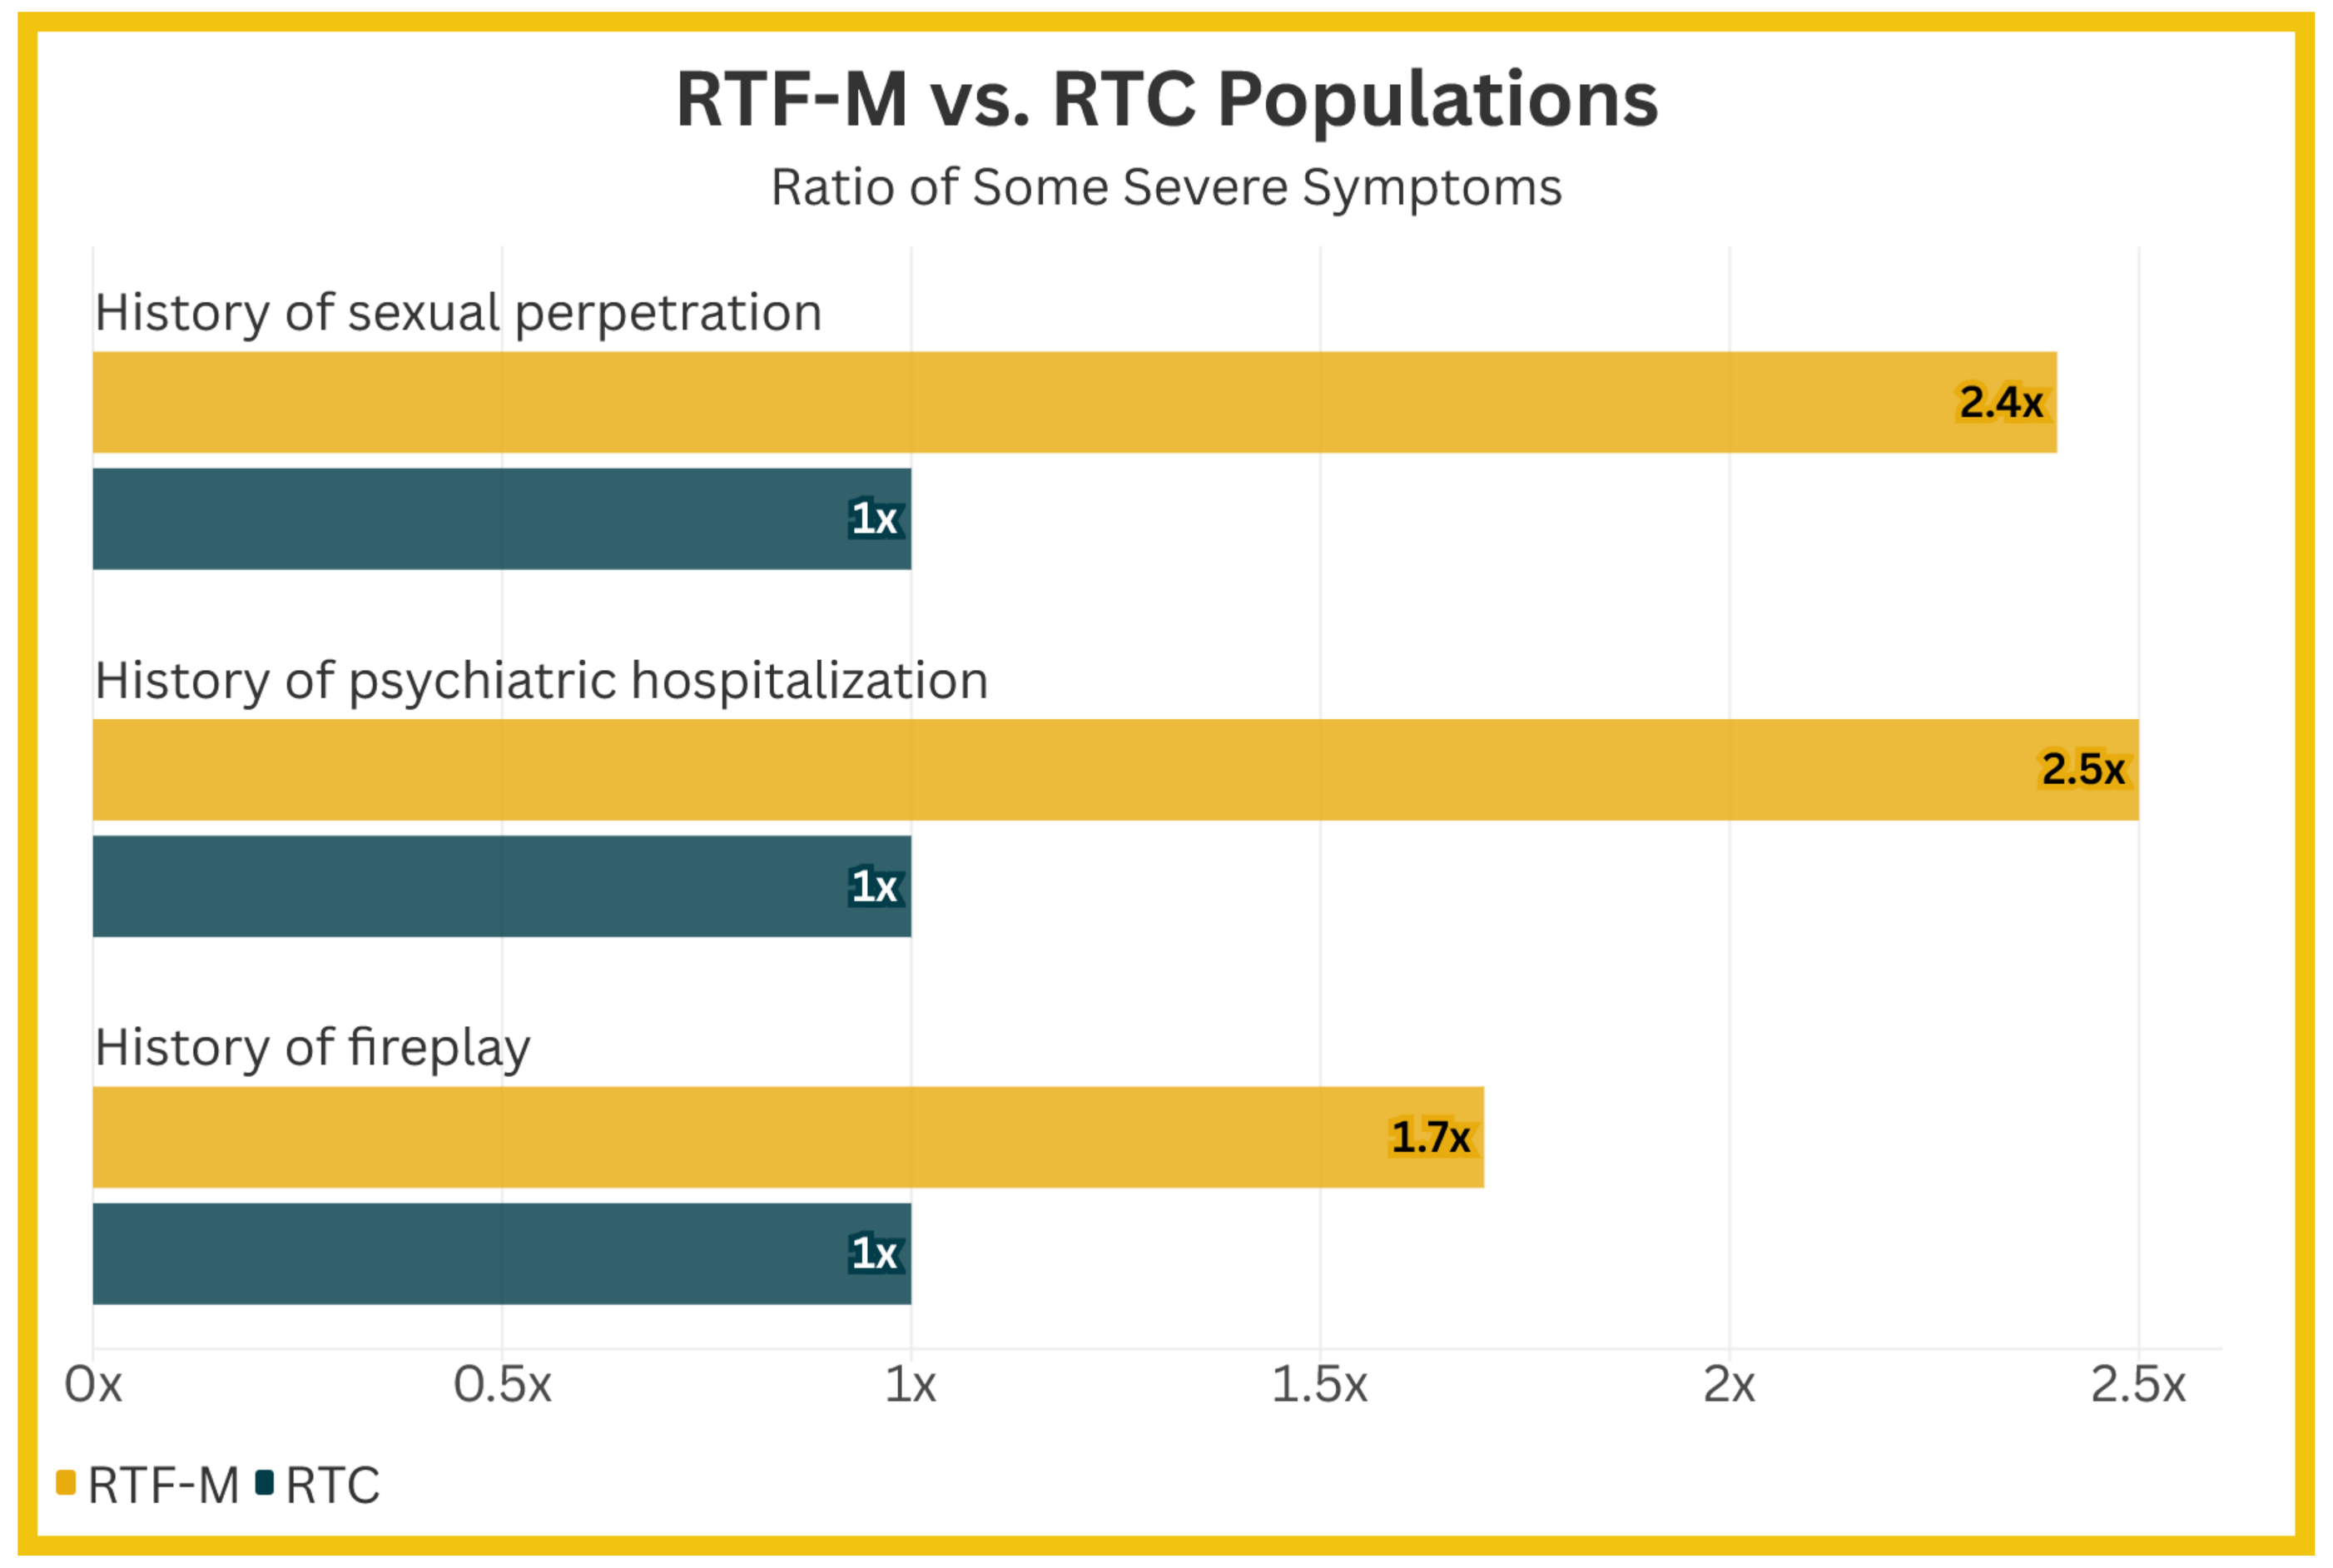

RTF-M vs. RTC Populations
Ratio of Some Severe Symptoms
History of sexual perpetration
2.4x
1x
History of psychiatric hospitalization
2.5x
1x
History of fireplay
1.7x
1x
0x
0.5x
1x
1.5x
2x
2.5x
RTF-M
RTC

REFERENCES

1. Baker AJ, Archer M, Curtis P. Youth characteristics associated with behavioral and mental health problems during the transition to residential treatment centers: the Odyssey Project population. Child Welfare. 2007;86(6):5-29.

2. Dale N, Baker AJ, Anastasio E, Purcell J. Characteristics of children in residential treatment in New York State. Child Welfare. 2007;86(1):5-27.

Higher needs are correlated with admission to the RTF-M vs to an RTC.

Severe maltreatment, high rates of pyschosis and hospitalization, extreme assaultiveness and suicidal behavior characterized the RTF population.

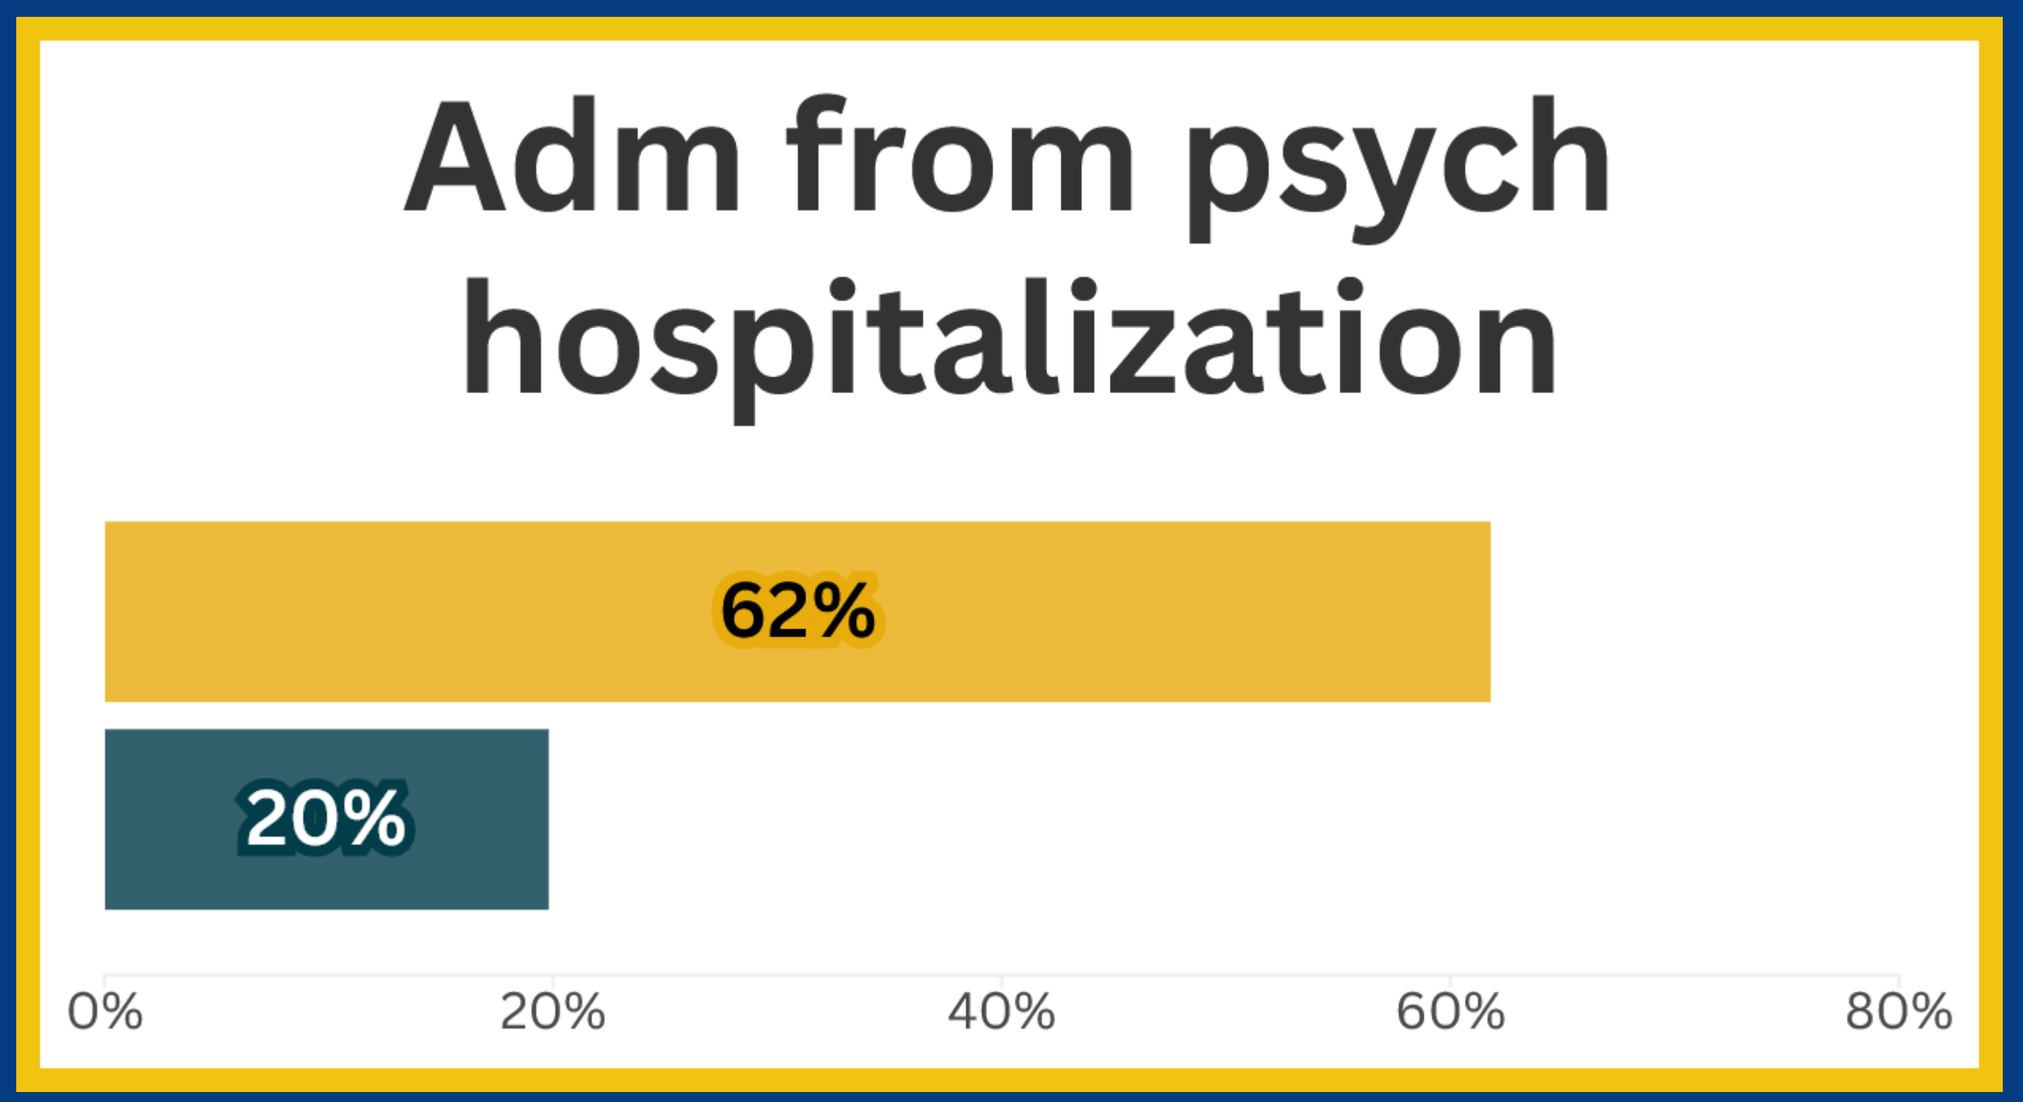

Adm from psych hospitalization
62%
20%
0%
20%
40%
60%
80%

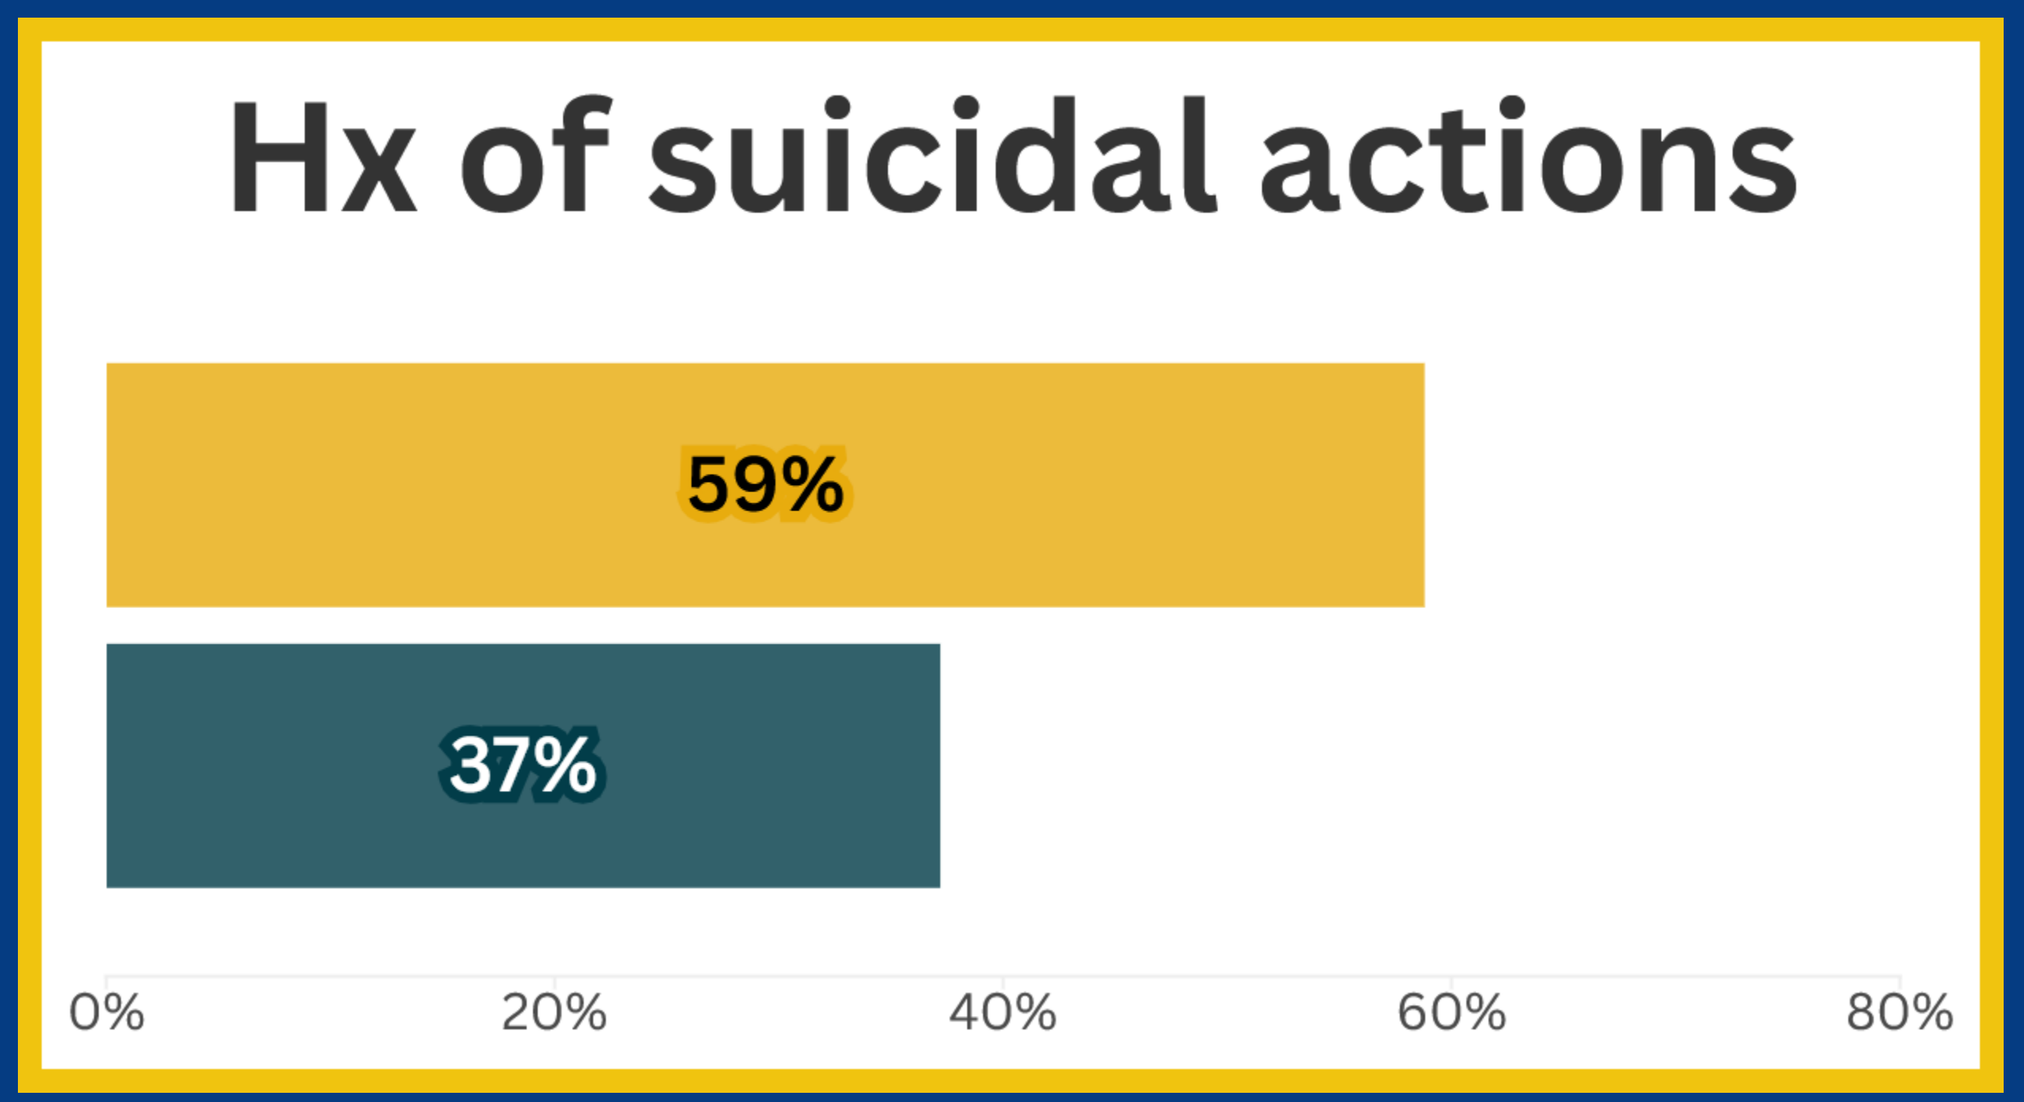

Hx of suicidal actions
59%
37%
0%
20%
40%
60%
80%

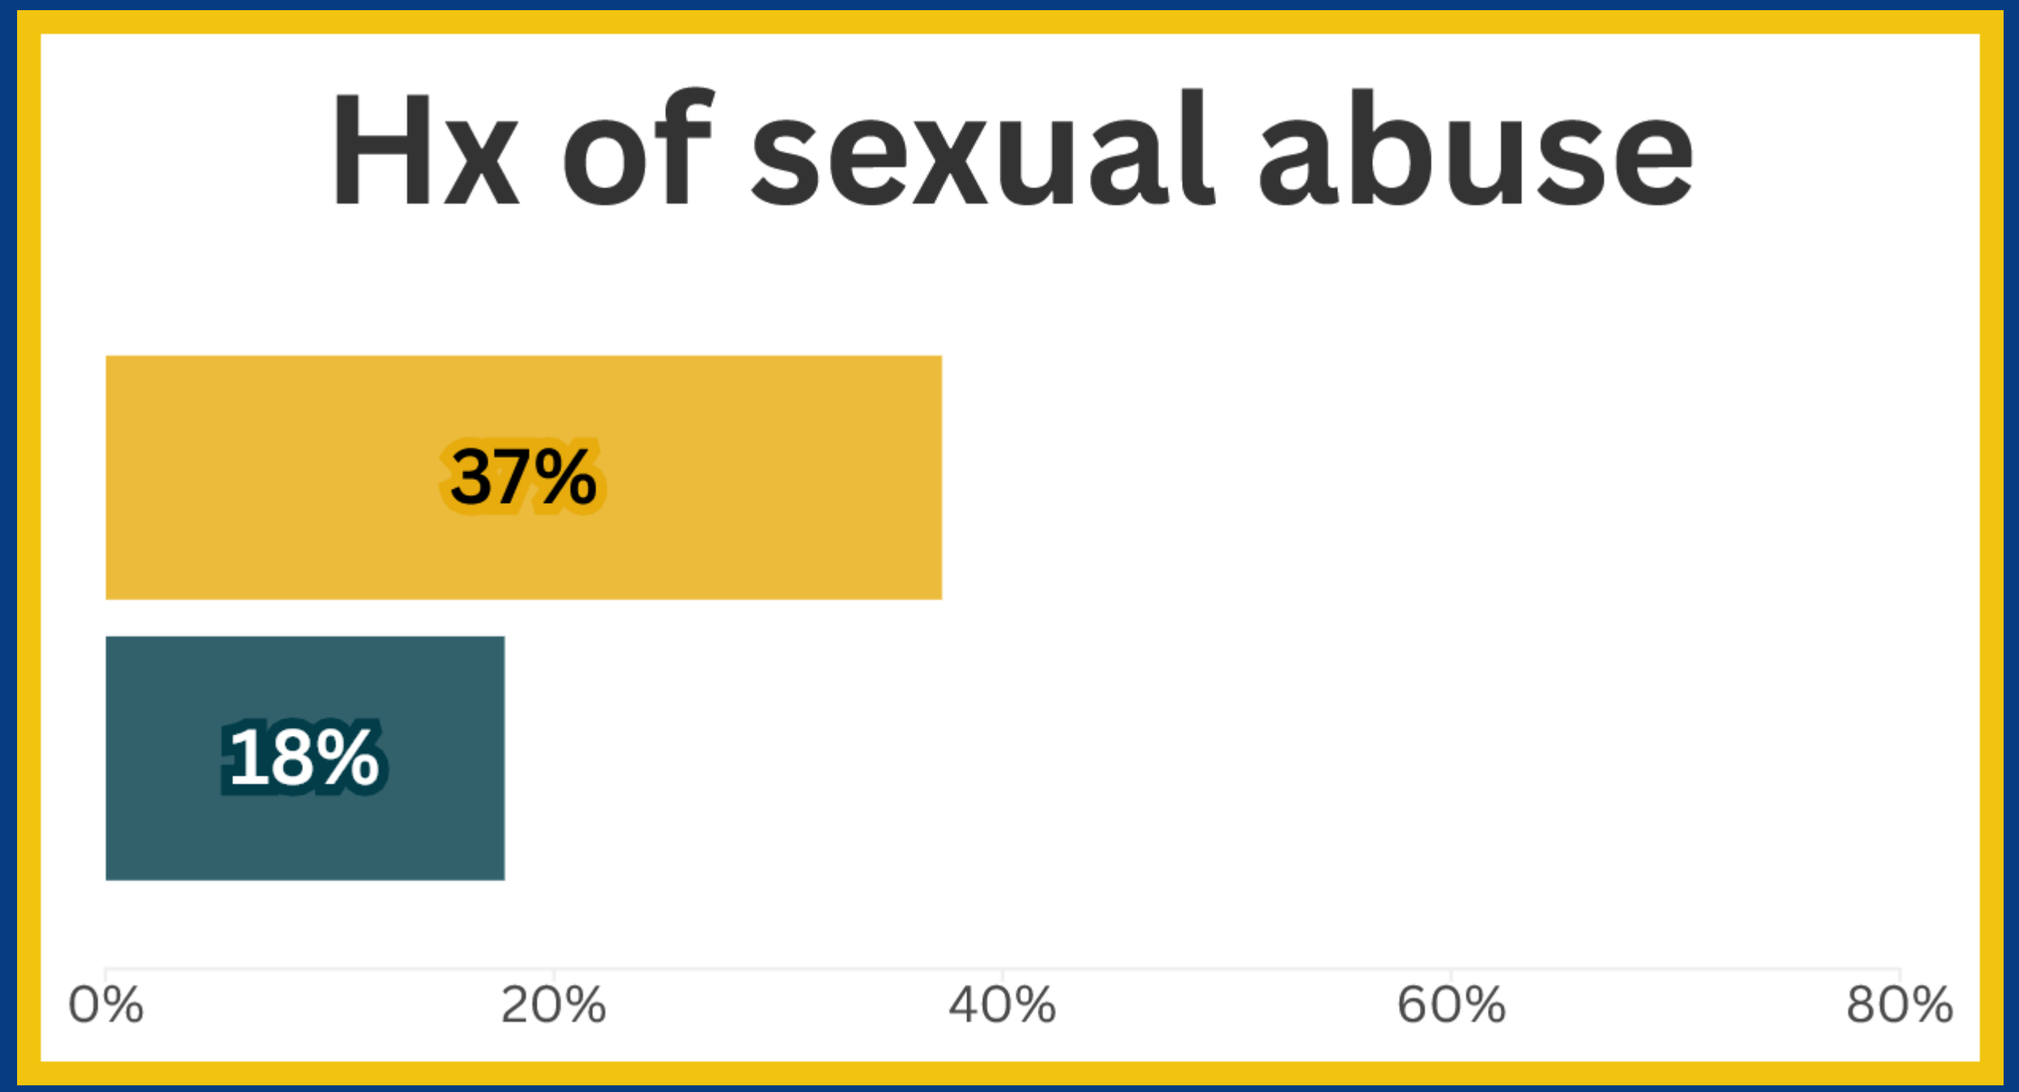

Hx of sexual abuse
37%
18%
0%
20%
40%
60%
80%

https://www.aichhorn.org/aichhome7-reports.html

Admitted From

	RTF-M	RTC (1995 - 2001)[1]
Mean age at admission	15.2	14.4
Proportion male	54.8%	72.3%
Home of parent or relative	3.6%	21.9%
Foster home	0%	4.5%
Therapeutic foster home	0.3%	0.9%
Group home	1.8%	5.7%
Emergency shelter	0.8%	3.5%
RTC	14.3%	18.8%
RTF	2.6%	-
Psychiatric hospitalization	61.8%	19.8%
Juvenile justice setting	14%	13.6%
Other / unknown	1%	11.4%

Admission History

	RTF-M	RTC (2001)[2]
Mean age at admission	15.2	13.9
Proportion male	54.8%	61.7%
History of child physical abuse	52.5%*	42.3%
History of child neglect	62.3%**	50.9%
History of child sexual abuse	37.3%***	17.8%
History of running away	61.8%*	51.7%
History of substance abuse	33.6%	41.6%*
History of fireplay	17.9%*	10.5%
Legal involvement prior to admission	35.3%	-
History of juvenile delinquency	38.6%	51.5%**
History of destruction of property	57.2%	-
History of physical aggression	90.6%	-
History of "major assaults"	85.9%	-
History of use of weapon	41.7%	-
History of "homicidal behavior"	41.3%	-
History of sexual perpetration	18.3%***	7.7%
History of sexual promiscuity	43.3%	-
History of enuresis and / or encopresis	14.1%	-
History of suicide actions	58.8%***	37.2%
History of suicide attempts	45.9%	-
History of self-harming behavior	53.2%	-
History of hallucinations (auditory and / or visual)	35.6%	-
History of psychotic symptoms and behaviors	44.1%	-
History of psychiatric hospitalization	96.7%***	39%
History of psychotropic medications	97.2%***	58.6%
Parent with history of psychiatric hospitalization	8.8%	7.9%
Parent with history of mental illness	30.7%	-
Parent with history of substance abuse	69.7%**	58.1%
Maternal substance abuse while pregnant	35.6%	-
Parent with history of imprisonment	23.2%	22.7%
Parent with history of antisocial / criminal behavior	28.1%	-
History of parental loss [i.e. parental death]	19.7%	-

* = p < .05 ** = p< .01 *** = p <.001